A Multicenter, Randomized, Double-Blind, Placebo-Controlled Trial of Nalbuphine ER Tablets for Uremic Pruritus

Vandana Mathur[1] and Thomas Sciascia[2]

[1]Mathur Consulting, CA; [2]Trevi Therapeutics, CT; USA

BACKGROUND

- A study of 73,000 United States dialysis patients reported that 60% have pruritus and that 30% are moderately to extremely bothered by it[1].
- Uremic pruritus is associated with significant decreases in quality of life (QoL), sleep, and greater use of IV antibiotics, ESA, and iron[1].
- The pathogenesis of uremic pruritus may involve an endogenous κ/μ opioid ligand ratio imbalance[2,3].
- Nalbuphine ER tablets (NAL) are an κ-opioid agonist/μ-opioid antagonist being developed for chronic pruritic conditions.

METHODS

- In this multicenter, randomized, double-blind trial 373 hemodialysis patients were randomized (1:1:1) to NAL 54 mg (NAL 54†; n = 128) or 108 mg (NAL 108†; n = 120) or placebo (n = 125) BID and treated for 8 weeks. Background antipruritic medications were allowed. Patients with pruritus related to other conditions (e.g., hepatic, malignancy, primary dermatologic condition) were excluded.
- The primary endpoint was the comparison between NAL 108 vs. placebo at Weeks 7-8 (Evaluation Period) on itching intensity using a Worst Itching Intensity Numerical Rating Scale (NRS, 0 [no itching] – 10 [worst possible itching]). Quality of life-related secondary endpoints included change from day 1 to the Evaluation Period in itching-related quality of life and itching-related sleep disruption.

RESULTS

- Demographics, dialysis adequacy, phosphorus, PTH, and antihistamine use were balanced (**Table 1**). The mean duration of itching was 3.2 years.

RESULTS, continued

- The primary efficacy endpoint was met. From a mean baseline worst-itch NRS of 6.9 (1.5), the mean NRS declined by 3.5 (2.4) in the NAL 108 group and by 2.8 (2.2) in the placebo group. This difference was statistically different (p = 0.017; **Figure 1**). There was no significant difference between NAL 54 and placebo.
- A significant separation between the NAL 108 group and placebo group was evident starting in the week following the blinded titration, with no apparent development of tolerance to the antipruritic effect during the 8-week treatment period (**Figure 2**).
- NRS scores increased during the off-drug washout period. The proportion of patients receiving antipruritic medications at baseline was similar in the treatment groups and there was no increase in the percentage of patients using antipruritic medications over time (**Figure 3**).
- The reduction in itching intensity was accompanied by trends in improvement in the quality-of-life measurement most proximate to itching intensity (bothersome nature of itching; p = 0.062 for NAL 108 vs. placebo) as well as in sleep disruption due to itching (p = 0.053 for NAL 108 vs placebo).
- The most common reason for discontinuing treatment in the active groups was opioid-type side effects (e.g., nausea and vomiting) that occurred during the forced titration period. One death occurred in the placebo group. The incidence of serious treatment-associated adverse events was 6.7%, 12.7%, and 15.4% in the NAL 108, NAL 54, and placebo groups, respectively.

Table 1. Baseline characteristics

	NAL 108 (n = 120)	NAL 54 (n = 128)	Placebo (n = 125)
Age (years)	55 (12)	55 (12)	56 (12)
Gender, % male	58	54	60
Race, White/Black	53/47	45/52	48/49
Hemodialysis duration (years)	4.7 (4.2)	4.8 (4.0)	4.5 (4.4)
Diabetes, %	50	56	48
PVD/PVD intervention, %	14/2	16/6	12/4
MI/ischemic HD intervention, %	8/15	12/21	17/7
Access [AVF/AVG/tunnel cath], %	73/18/9	75/15/8	70/18/11
Urea reduction ratio/Kt/V	74/1.5	74/1.6	75/1.6
iPTH (pg/mL)	452 (455)	382 (318)	464 (590)
Phosphate (mg/dL)	5.6 (1.5)	5.4 (1.8)	5.7 (1.0)

Figure 1. Change in itching

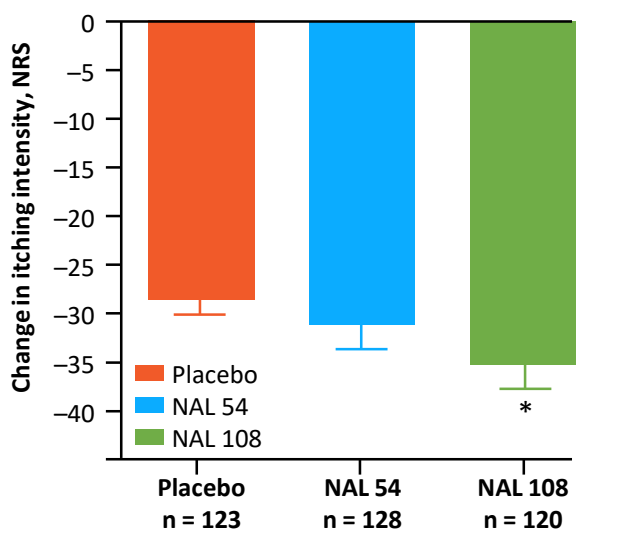
Change in itching intensity, NRS
0
−5
−10
−15
−20
−25
−30
−35
−40
Placebo
NAL 54
NAL 108
*
Placebo n = 123
NAL 54 n = 128
NAL 108 n = 120

* p = 0.017 vs. placebo (observed data based on mixed effect model repeated measurement).

Figure 2. Itching reduction over time

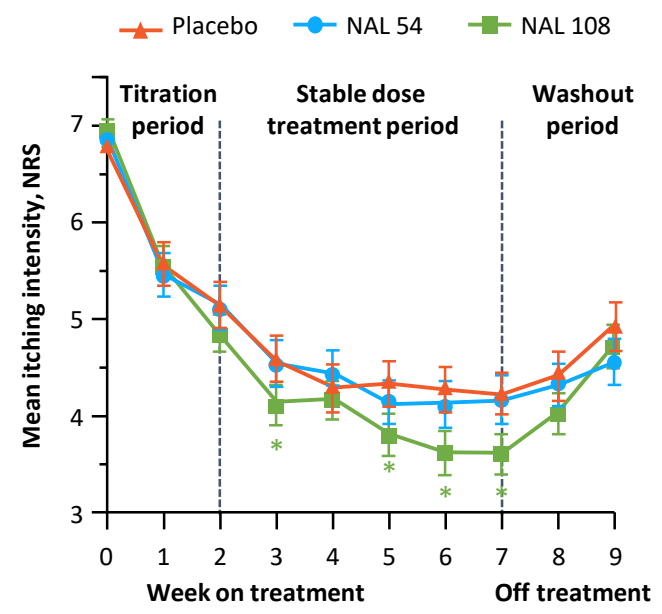
Placebo
NAL 54
NAL 108
Titration period
Stable dose treatment period
Washout period
Mean itching intensity, NRS
3
4
5
6
7
0 1 2 3 4 5 6 7 8 9
Week on treatment
Off treatment

Data depicted are means (SEM). * p < 0.05 vs. placebo.

Figure 3. Antipruritic medication usage

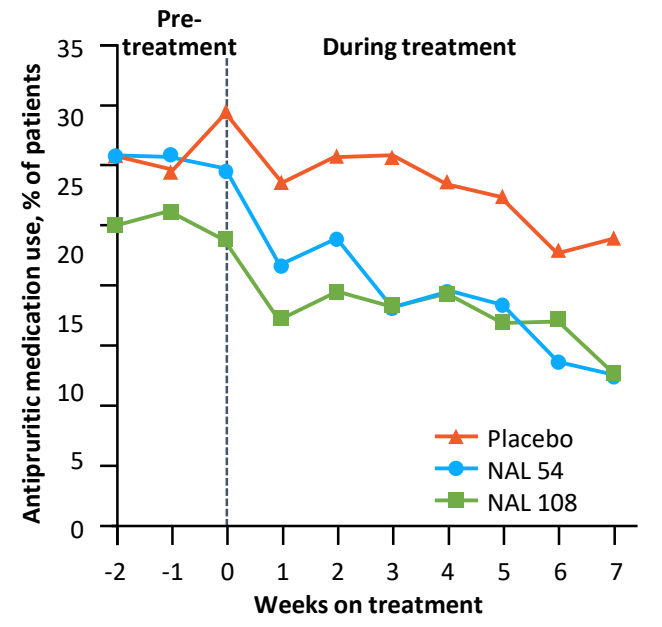
Pre-treatment
During treatment
Antipruritic medication use, % of patients
0
5
10
15
20
25
30
35
-2 -1 0 1 2 3 4 5 6 7
Weeks on treatment
Placebo
NAL 54
NAL 108

CONCLUSION

- The trial met its primary endpoint, demonstrating a significant reduction in itch intensity in the NAL 108 group vs. placebo in hemodialysis patients with moderate and severe uremic pruritus receiving background antipruritic drugs such as antihistamines and topical corticosteroids.
- The effect of NAL 108 was evident within 1 week following titration and was durable for the full 8-week treatment period.
- This large randomized trial in uremic pruritus demonstrated the efficacy of nalbuphine ER tablets for one of the most distressing complications of end-stage renal disease.

ACKNOWLEDMENT

Funding toward this study was received from Trevi Therapeutics. Writing/editorial support was provided by Excerpta Medica, funded by Trevi Therapeutics.

REFERENCES

1. Ramakrishnan K et al. Int J Nephrol Renovasc Dis 2013;7:1-12.
2. Kumagai H et al. In: Itch Basic Mechanisms and Therapy 2004.
3. Wang H & Yosipovitch G. Int J Dermatol 2010;49:1-11.

†Previous publications used the molecular weight including salts (60 mg and 120 mg), whereas the doses described in this poster are for the molecular weight of just the active drug.

ePoster presented at the 2020 Fall Clinical Dermatology Conference, Wynn, Las Vegas, NV, USA, Oct 29 – Nov 1, 2020